A NOVEL MULTI-INSERT MVA-BN BASED RSV VACCINE PROVIDES IMPROVED PROTECTION IN MICE AND COTTON RATS

ARIANE VOLKMANN[1], KATHRIN ENDT[1], CÉDRIC CHEMINAY[1,A], MARKUS KALLA[1], ROBIN STEIGERWALD[1], SONIA T. WENNIER[1], BRIAN E. GILBERT[2], PAUL CHAPLIN[3]

[1]BAVARIAN NORDIC GMBH, FRAUNHOFERSTR. 13, 82152 MARTINSRIED, GERMANY [2]BAYLOR COLLEGE OF MEDICINE, HOUSTON, TX77030-3498, USA [A]CURRENT ADDRESS: VIRATHERAPEUTICS GMBH, EXLGASSE 20A, A-6020 INNSBRUCK, AUSTRIA, [3]BAVARIAN NORDIC A/S, HEJRESKOVVEJ 10A, 3490 KVISTGÅRD, DENMARK

BACKGROUND

MVA-BN-RSV Vaccine Design

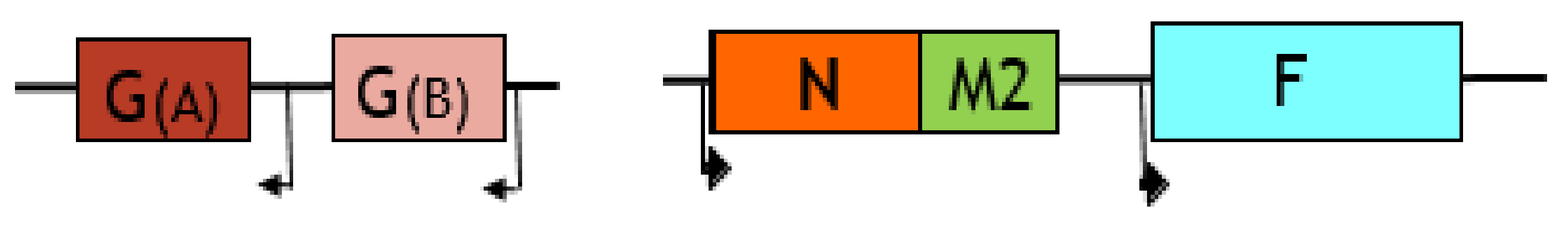
G(A)
G(B)
N
M2
F

RSV can cause severe respiratory tract infections not only in infants, but also in older adults. No licensed vaccine against RSV is currently available. Recent attempts relying solely on the RSV fusion- (F) protein failed. We designed MVA based RSV vaccines that express either RSV F only, or additional RSV antigens and evaluated the role of these RSV antigens in vaccine efficacy.

The lead candidate MVA-BN-RSV is depicted above.

METHODS

BALB/c mice or cotton rats were vaccinated on Days 0 & 21 with 1×10^8 $TCID_{50}$ of MVA-BN based constructs expressing either RSV F only, RSV F and G (A subtype), or RSV F, G (A & B subtype), N and M2, i.e. MVA-BN-RSV. RSV-A2 was used as a positive control, Tris buffered saline (TBS, non-vaccinated) as negative control.

Two weeks later, animals were intranasally infected with RSV A2 and viral load was determined by plaque assay or RT-qPCR 4 days post infection. Enhanced respiratory disease induced by FI-RSV was evaluated in terms of IL-5 and eosinophils. The expression of pre-F versus post-F by HeLa cells infected with recombinant MVA-BN was determined by flow cytometry.

MVA-BN based RSV vaccines are immunogenic in mice

- All constructs induced comparable levels of antibodies, including neutralizing antibodies (latter not shown)
- The addition of more RSV antigens increased the magnitude of T cell responses, especially against internal proteins, while mimicking the composition of RSV-A2

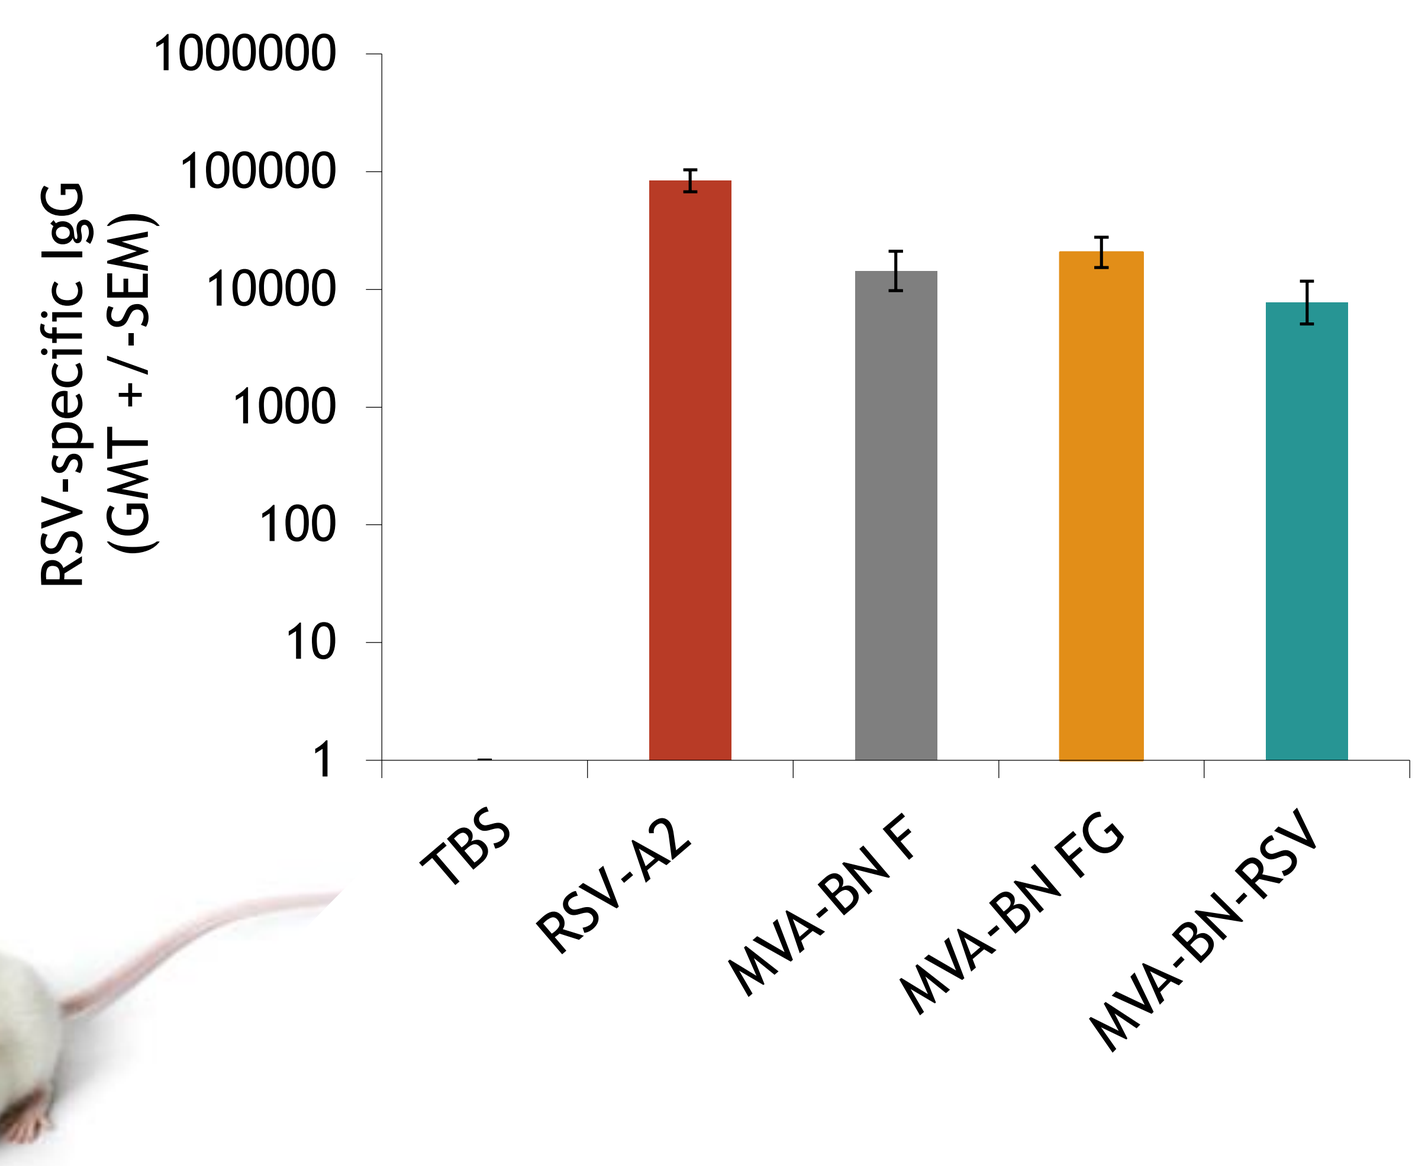
RSV-specific IgG (GMT +/-SEM)
1000000
100000
10000
1000
100
10
1
TBS
RSV-A2
MVA-BN F
MVA-BN FG
MVA-BN-RSV

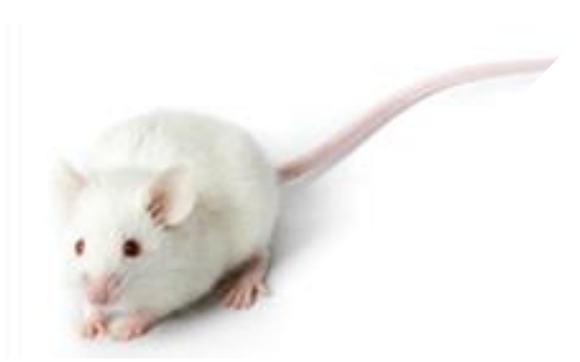

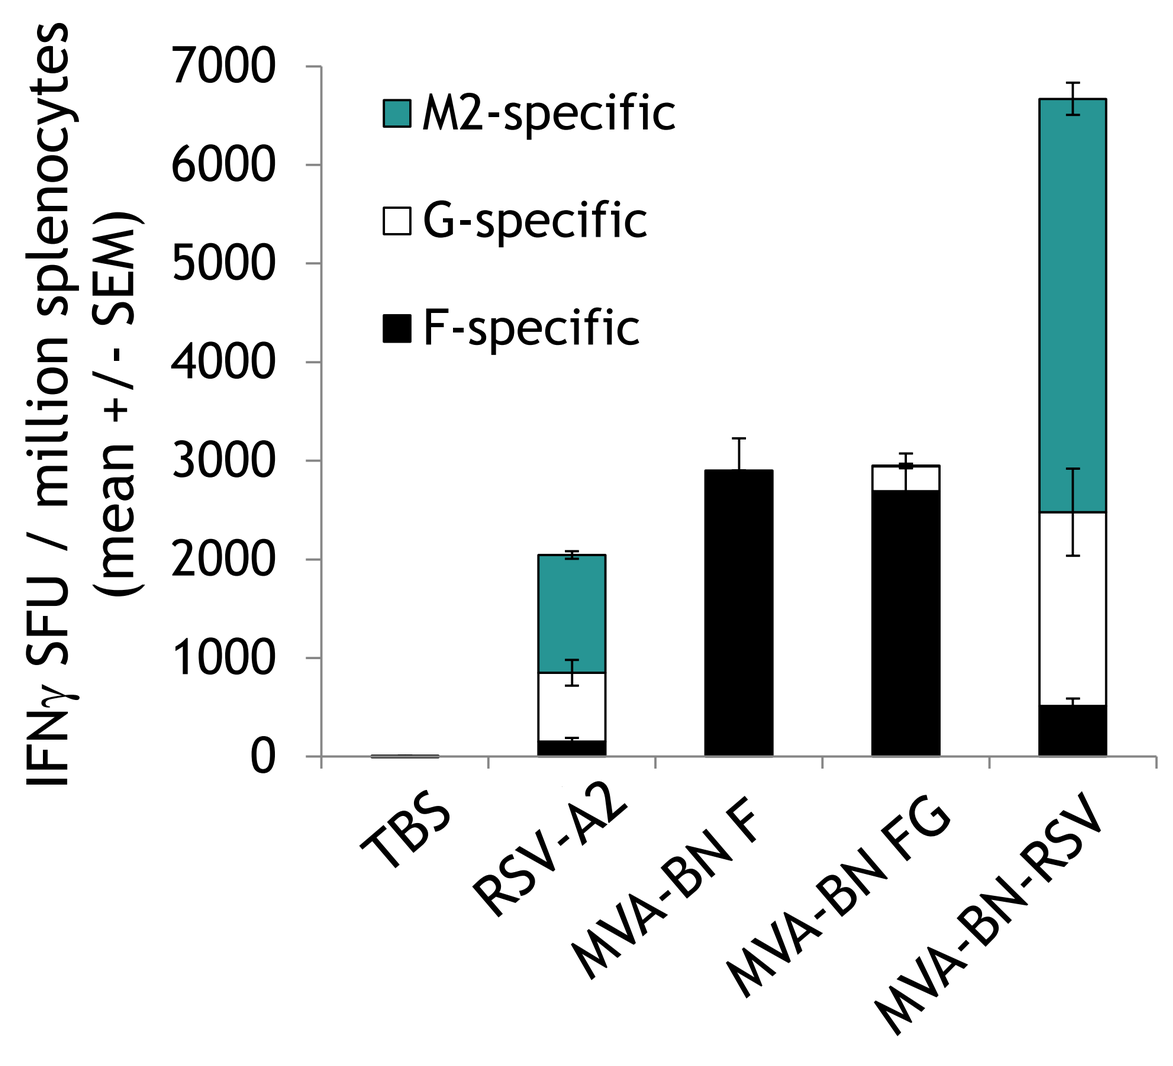
IFNγ SFU / million splenocytes (mean +/- SEM)
7000
6000
5000
4000
3000
2000
1000
0
M2-specific
G-specific
F-specific
TBS
RSV-A2
MVA-BN F
MVA-BN FG
MVA-BN-RSV

Additional RSV antigens increase vaccine efficacy in mice

- Infectious RSV was still detectable in 3 of 5 mice vaccinated with MVA-BN-F, whereas the addition of G led to total clearance in lung
- Improved efficacy with the expression of additional antigens in the sensitive RT-qPCR
- Protection afforded by MVA-BN-RSV was comparable to that induced by RSV exposure

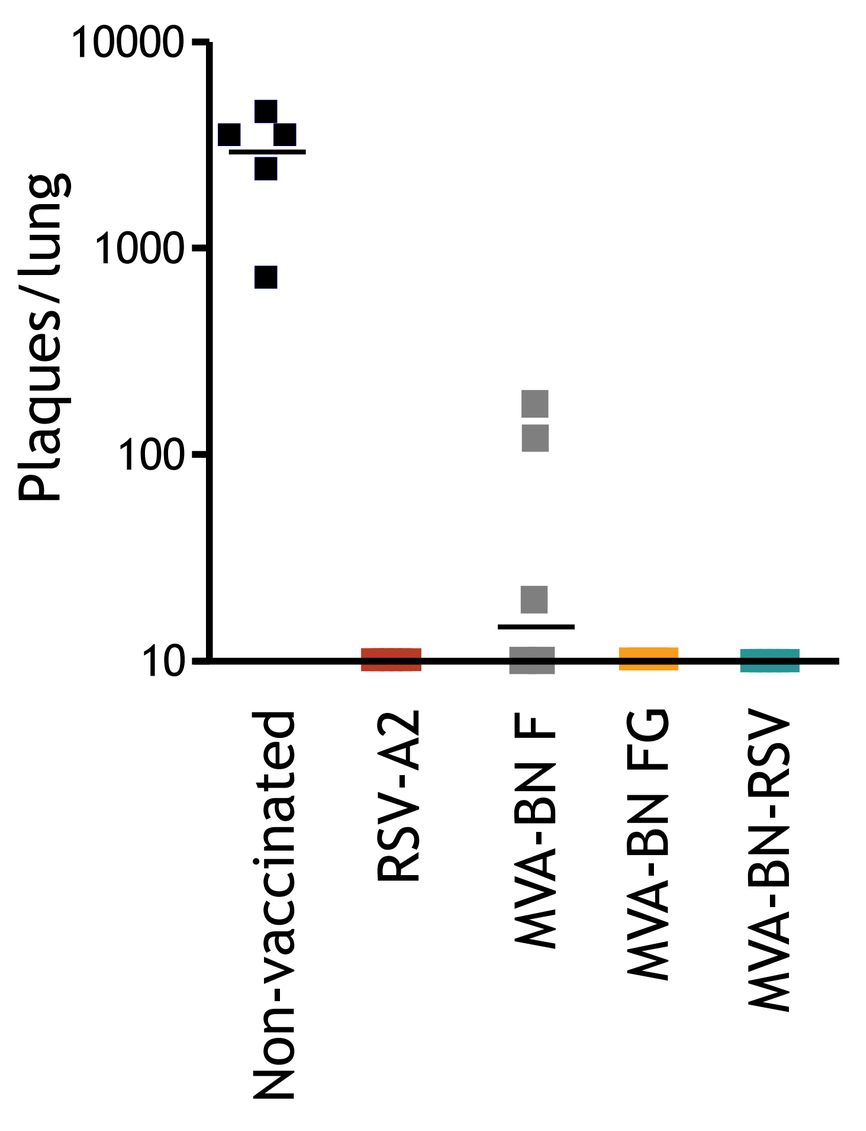
Plaques/lung
10000
1000
100
10
Non-vaccinated
RSV-A2
MVA-BN F
MVA-BN FG
MVA-BN-RSV

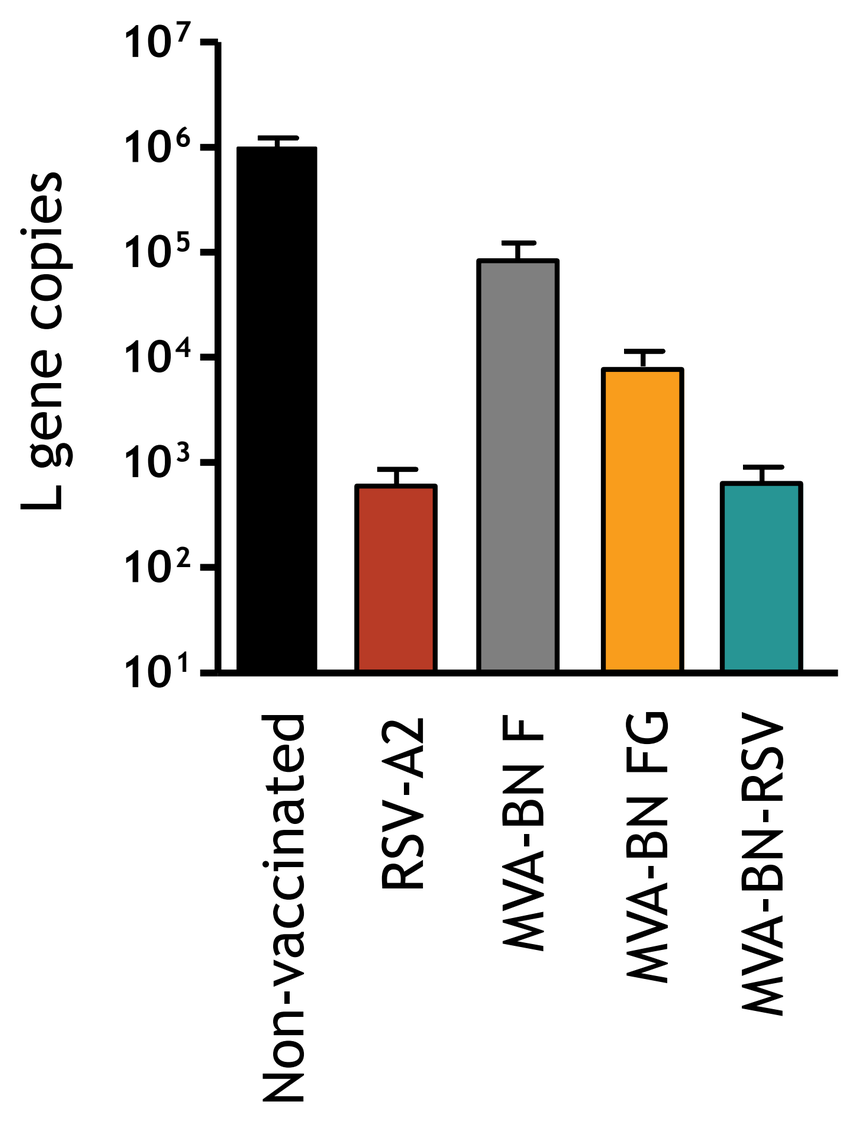
L gene copies
10^7
10^6
10^5
10^4
10^3
10^2
10^1
Non-vaccinated
RSV-A2
MVA-BN F
MVA-BN FG
MVA-BN-RSV

MVA-BN-RSV does not induce enhanced respiratory disease

- In contrast to formalin inactivated (FI) RSV, MVA-BN-RSV does not induce elevated serum IL-5 or eosinophil infiltration in the lung evaluated by cytospin and histology 4 days after RSV infection.

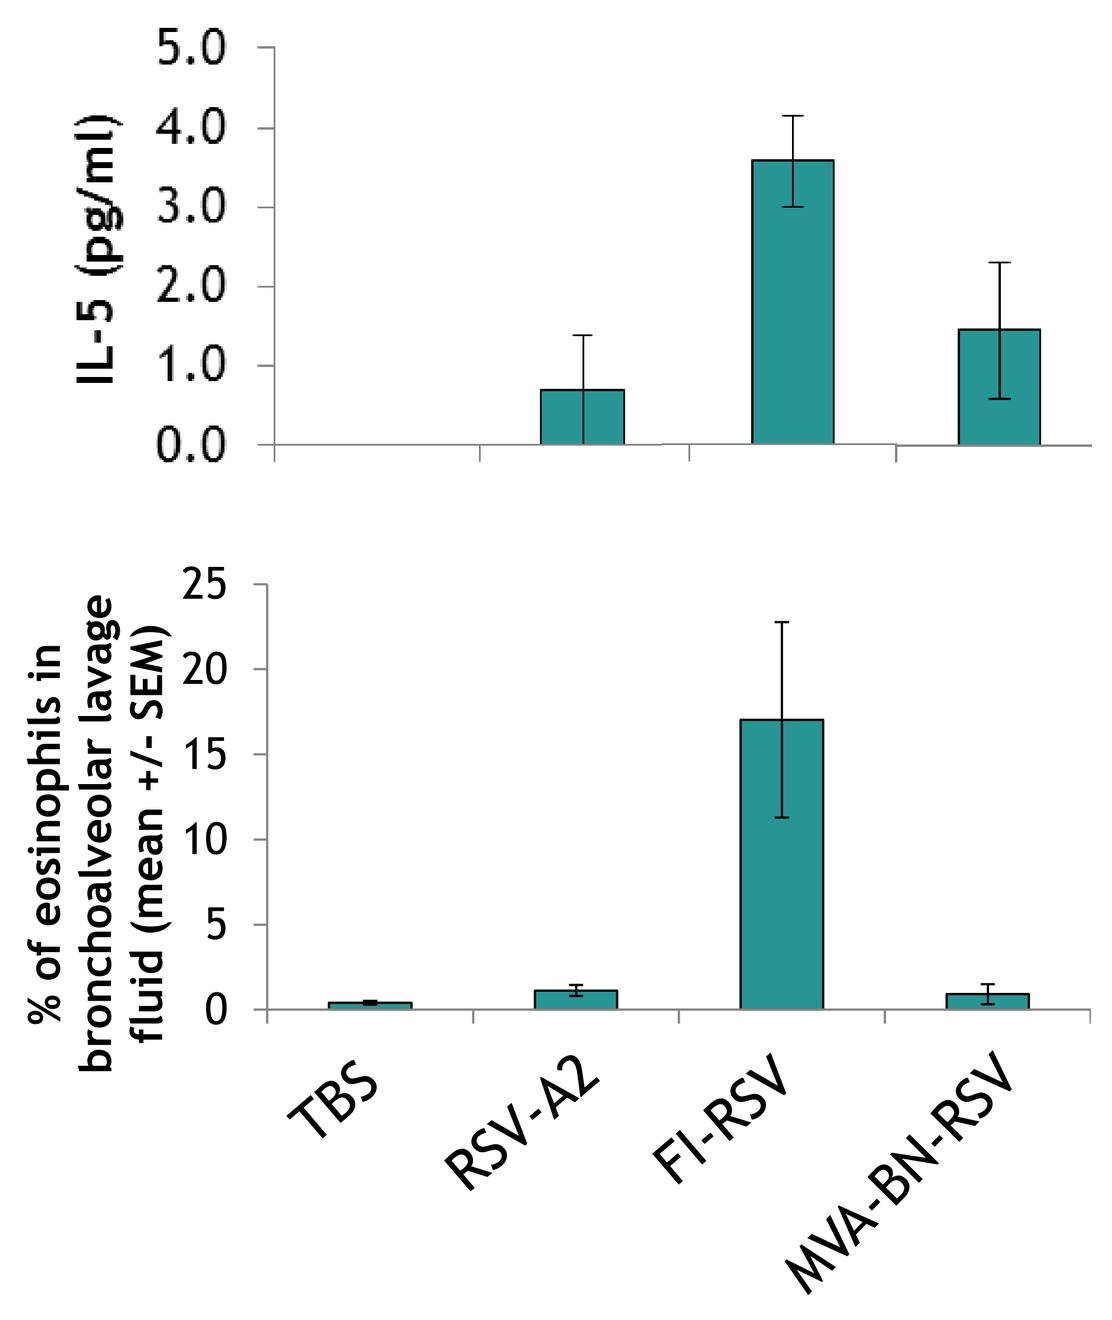
IL-5 (pg/ml)
5.0
4.0
3.0
2.0
1.0
0.0
% of eosinophils in bronchoalveolar lavage fluid (mean +/- SEM)
25
20
15
10
5
0
TBS
RSV-A2
FI-RSV
MVA-BN-RSV

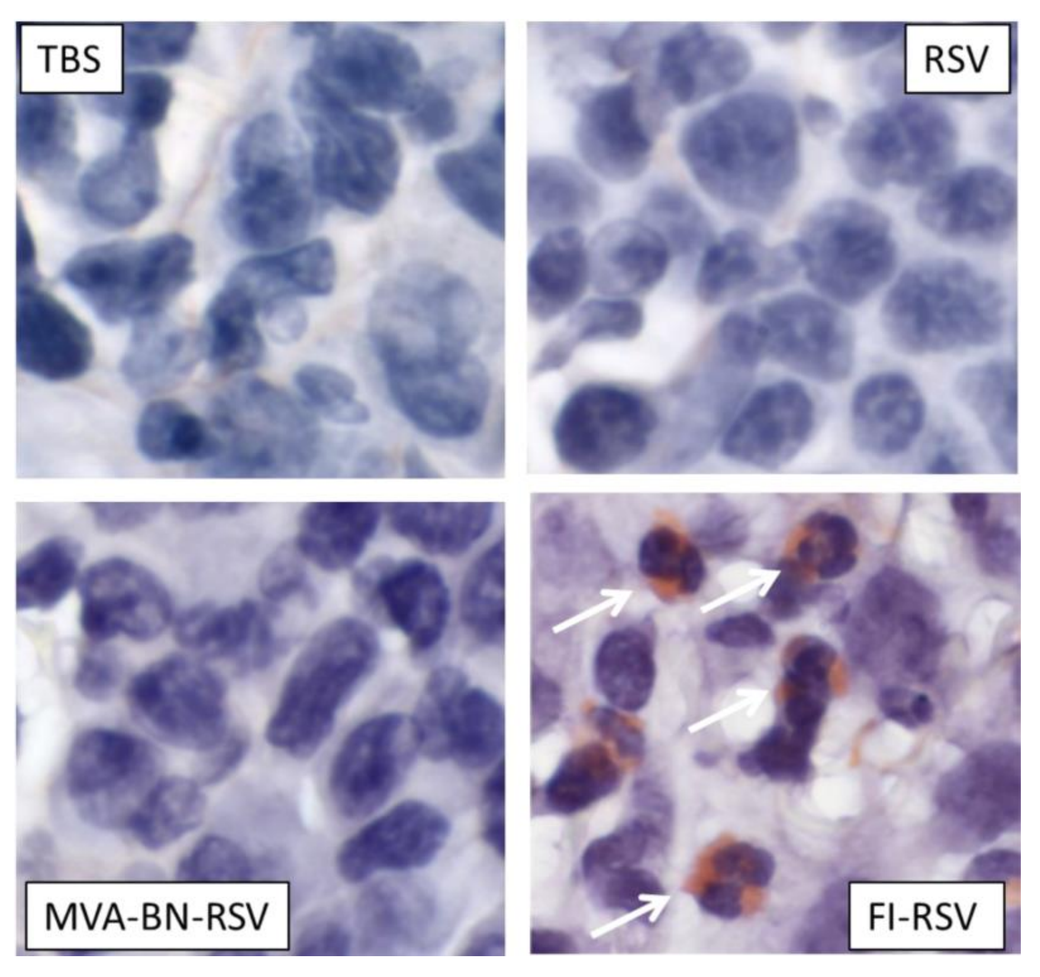
TBS
RSV
MVA-BN-RSV
FI-RSV

Additional RSV antigens increase vaccine efficacy in cotton rats

- Additional RSV antigens in MVA-BN-RSV led to higher levels of RSV-A and RSV-B neutralizing antibodies
- RSV-A neutralizing antibodies were comparable to those induced by RSV-A2, while RSV-B neutralizing antibodies were increased, indicating a benefit of the B subtype G protein encoded by MVA-BN-RSV
- Protective efficacy was increased by additional RSV antigens and comparable to that afforded by the RSV-A2 control

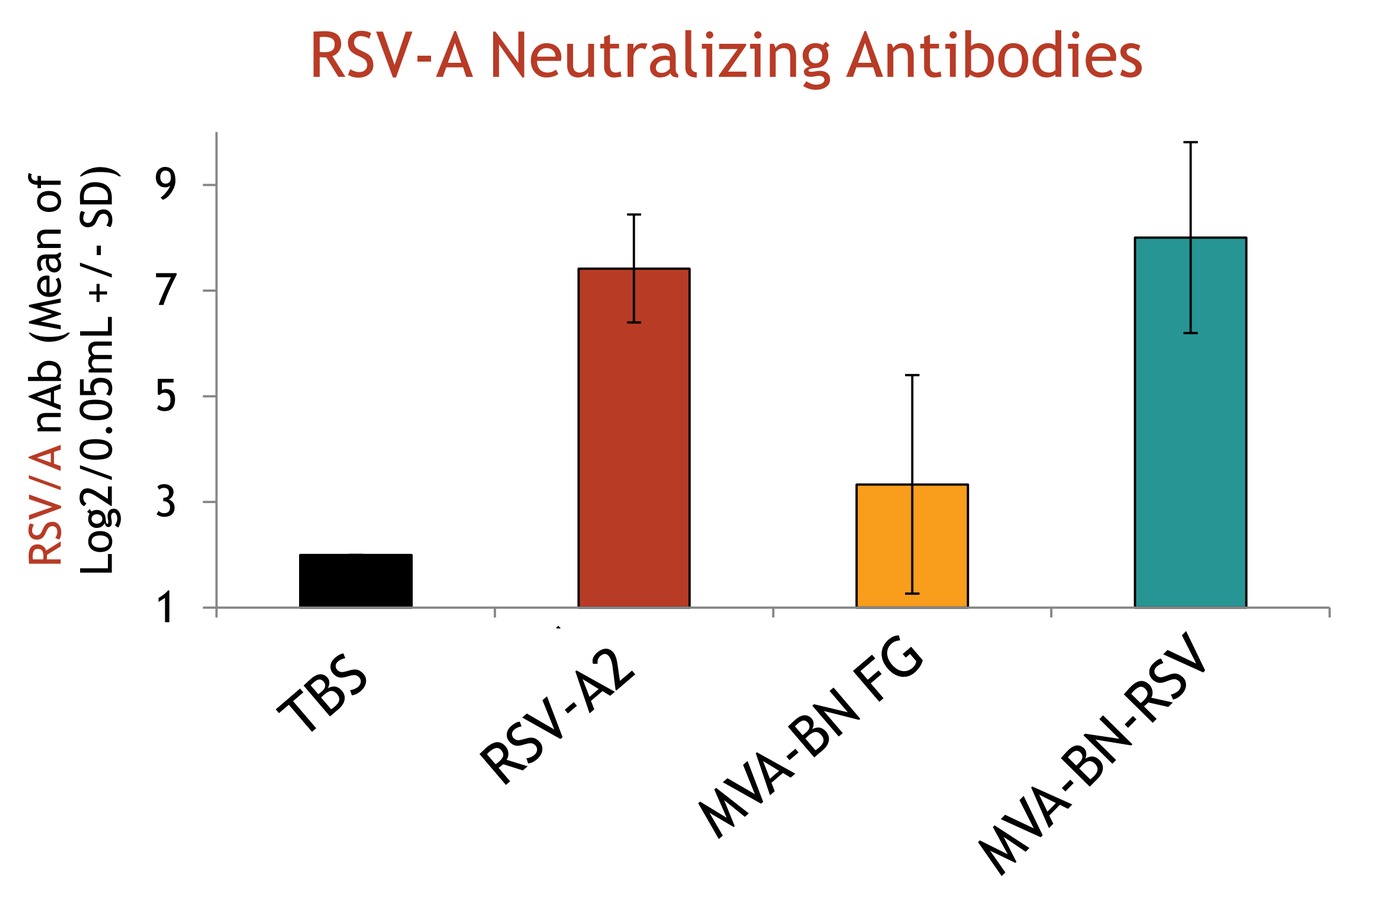
RSV-A Neutralizing Antibodies
RSV/A nAb (Mean of Log2/0.05mL +/- SD)
9
7
5
3
1
TBS
RSV-A2
MVA-BN FG
MVA-BN-RSV

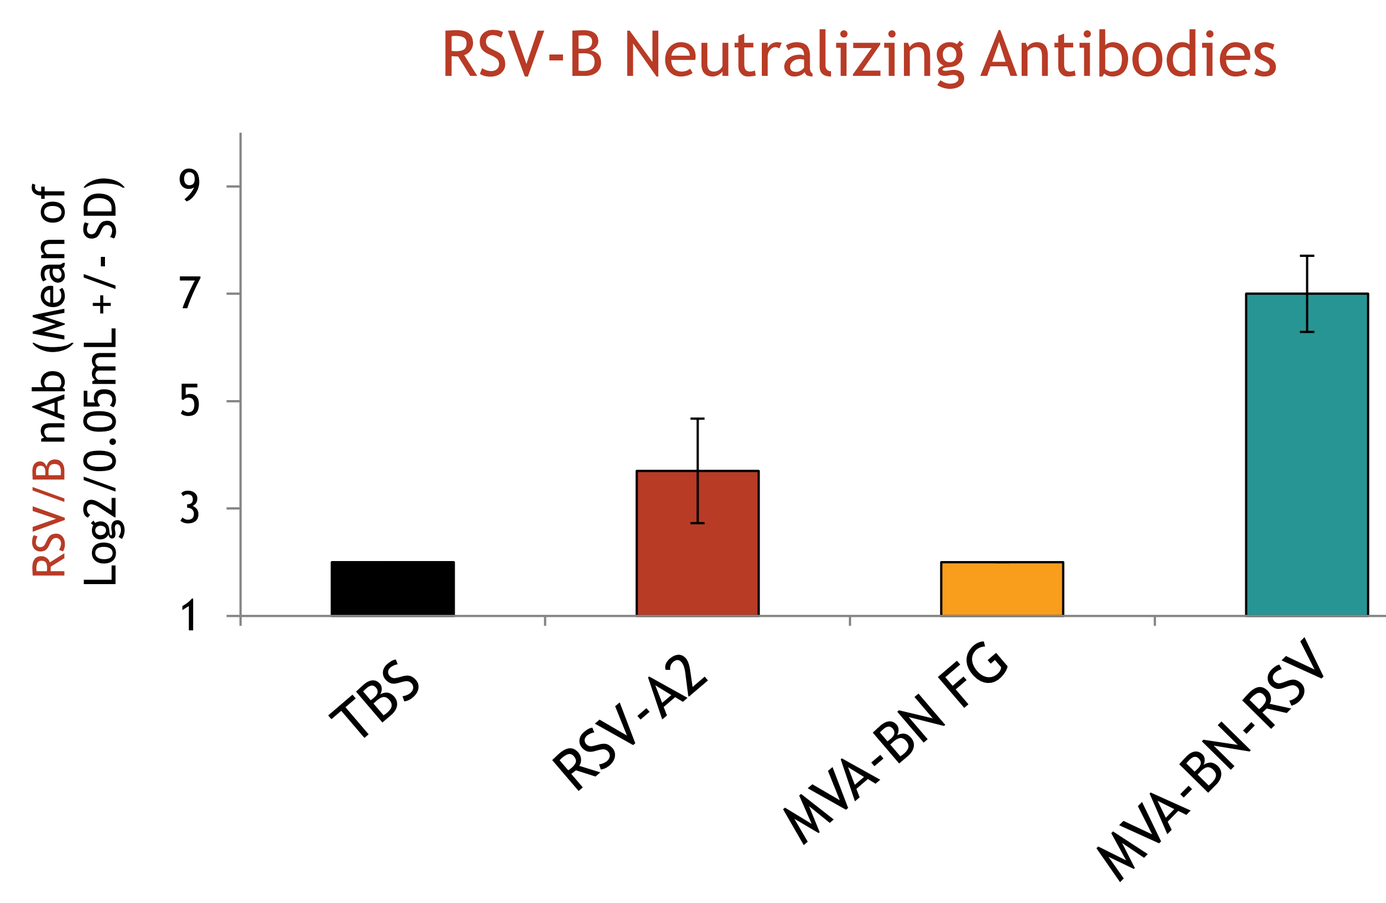
RSV-B Neutralizing Antibodies
RSV/B nAb (Mean of Log2/0.05mL +/- SD)
9
7
5
3
1
TBS
RSV-A2
MVA-BN FG
MVA-BN-RSV

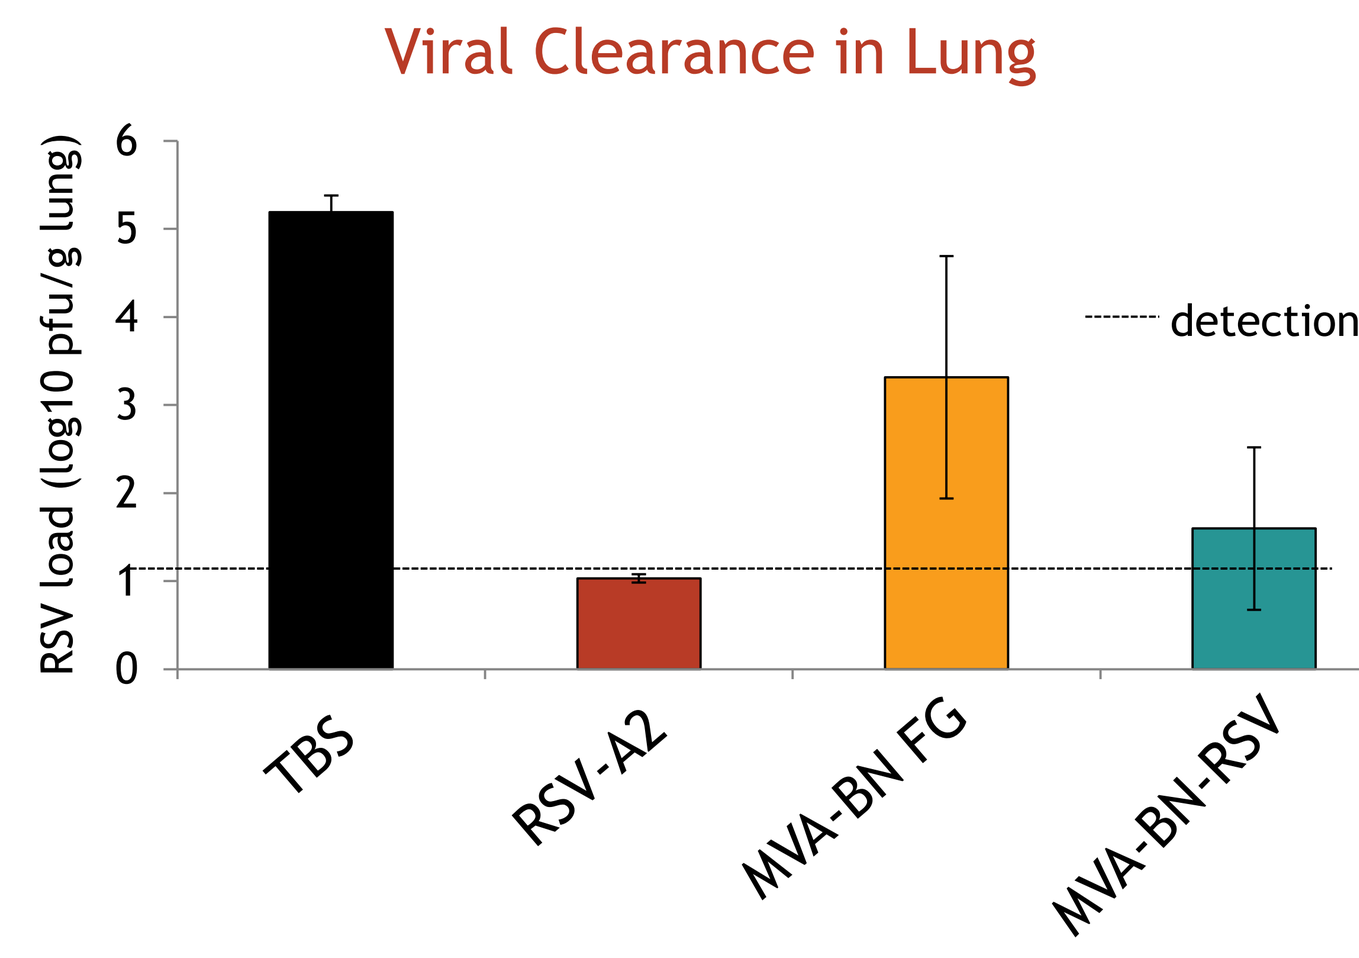
Viral Clearance in Lung
RSV load (log10 pfu/g lung)
6
5
4
3
2
1
0
detection limit
TBS
RSV-A2
MVA-BN FG
MVA-BN-RSV

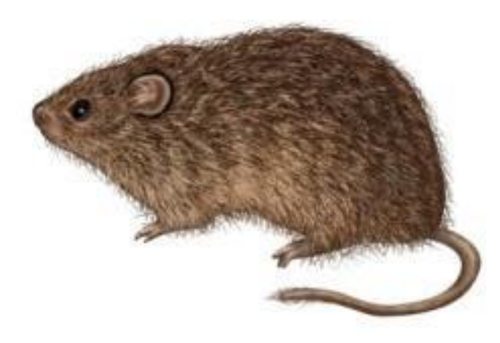

MVA-BN-RSV`s native F antigen displays both pre- and post-F conformations

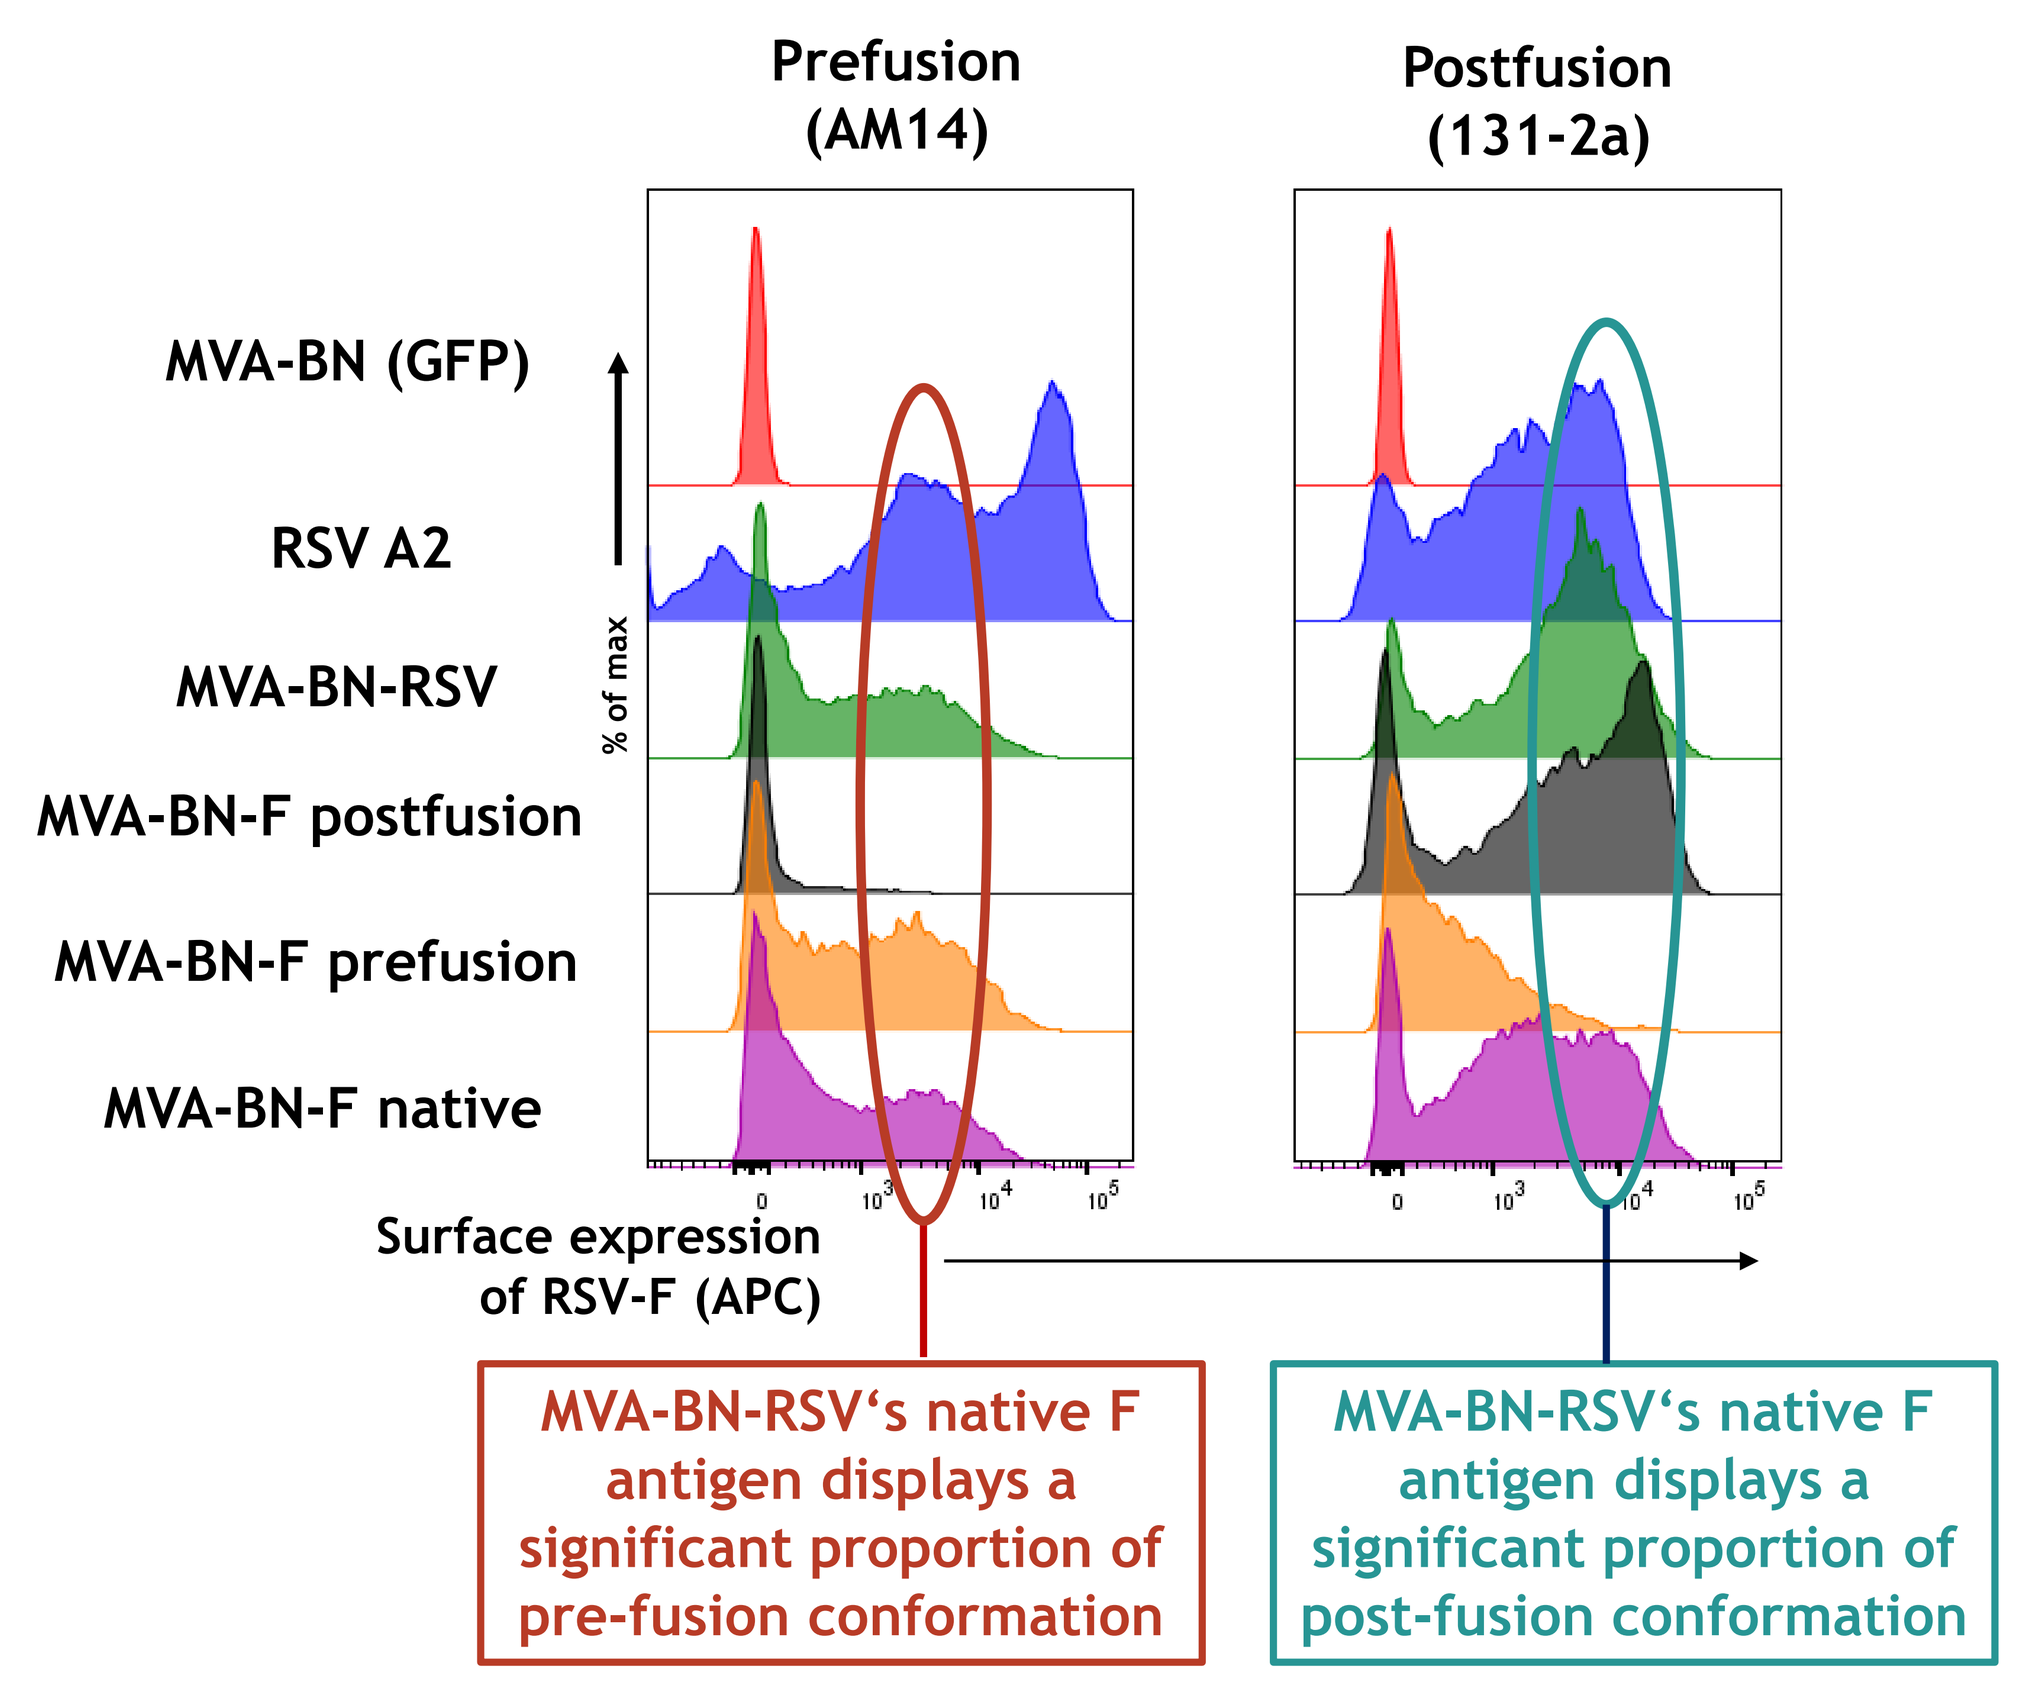
Prefusion (AM14)
Postfusion (131-2a)
MVA-BN (GFP)
RSV A2
MVA-BN-RSV
MVA-BN-F postfusion
MVA-BN-F prefusion
MVA-BN-F native
% of max
Surface expression of RSV-F (APC)
MVA-BN-RSV's native F antigen displays a significant proportion of pre-fusion conformation
MVA-BN-RSV's native F antigen displays a significant proportion of post-fusion conformation

CONCLUSIONS

- Expression of multiple RSV antigens increased the immunogenicity and efficacy of an MVA based RSV vaccine compared to the expression of F alone
- Limited efficacy of the F-only construct is not due to a lack of the pre-F confirmation as shown by the presence of both pre- and post-F in MVA-BN-F infected cells
- MVA-BN-RSV encoding F, G and M2/N was shown to be as immunogenic and efficacious as RSV in two different animal challenge models, without inducing enhanced disease
- Phase 1 and 2 clinical trials recently confirmed the favorable safety profile and immunogenicity of MVA-BN-RSV

Acknowledgements
The cotton rat studies were supported by the National Institutes of Health, Contract No. HHSN272201000004I

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

BAVARIAN NORDIC